OCTOBER 11-12, 2024 MILAN, ITALY DeltaCure 3rd International Meeting

P48

Poster for you

Adenine Base Editing of Hepatitis B surface antigen potently inhibits HDV

A. Kumar[1,2,3], E. Combe[1,2,3], E.M. Smekalova[4], S. Dejene[4], C-Y. Chen[4], L. Mougené[1,2,3], M.G. Martinez[1,2,3], G. Ciaramella[4], M.S. Packer[4], **B. Testoni**[1,2,3], F. Zoulim[1,2,3,4,5]

[1]INSERM U1052, CNRS UMR-5286,Cancer Research Center of Lyon, Lyon, France; [2]University of Lyon, Université Claude-Bernard (UCBL), Lyon, France; [3]The Lyon Hepatology Institute EVEREST, Lyon, France; [4]Beam Therapeutics, Cambridge, MA, USA; [5]Department of Hepatology, Croix Rousse hospital, Hospices Civils de Lyon, Lyon, France

Introduction

Scientific Background

- Chronic Hepatits B virus (HBV) and Hepatitis D virus (HDV) infections represent serious health burden
- HBsAg is the common structural component of HBV and HDV
- HBsAg loss is considered as a desired endpoint of anti-HBV therapy, but is rarely achieved by current treatments
- Lowering HBsAg secretion is also being explored as an approach to inhibit HDV

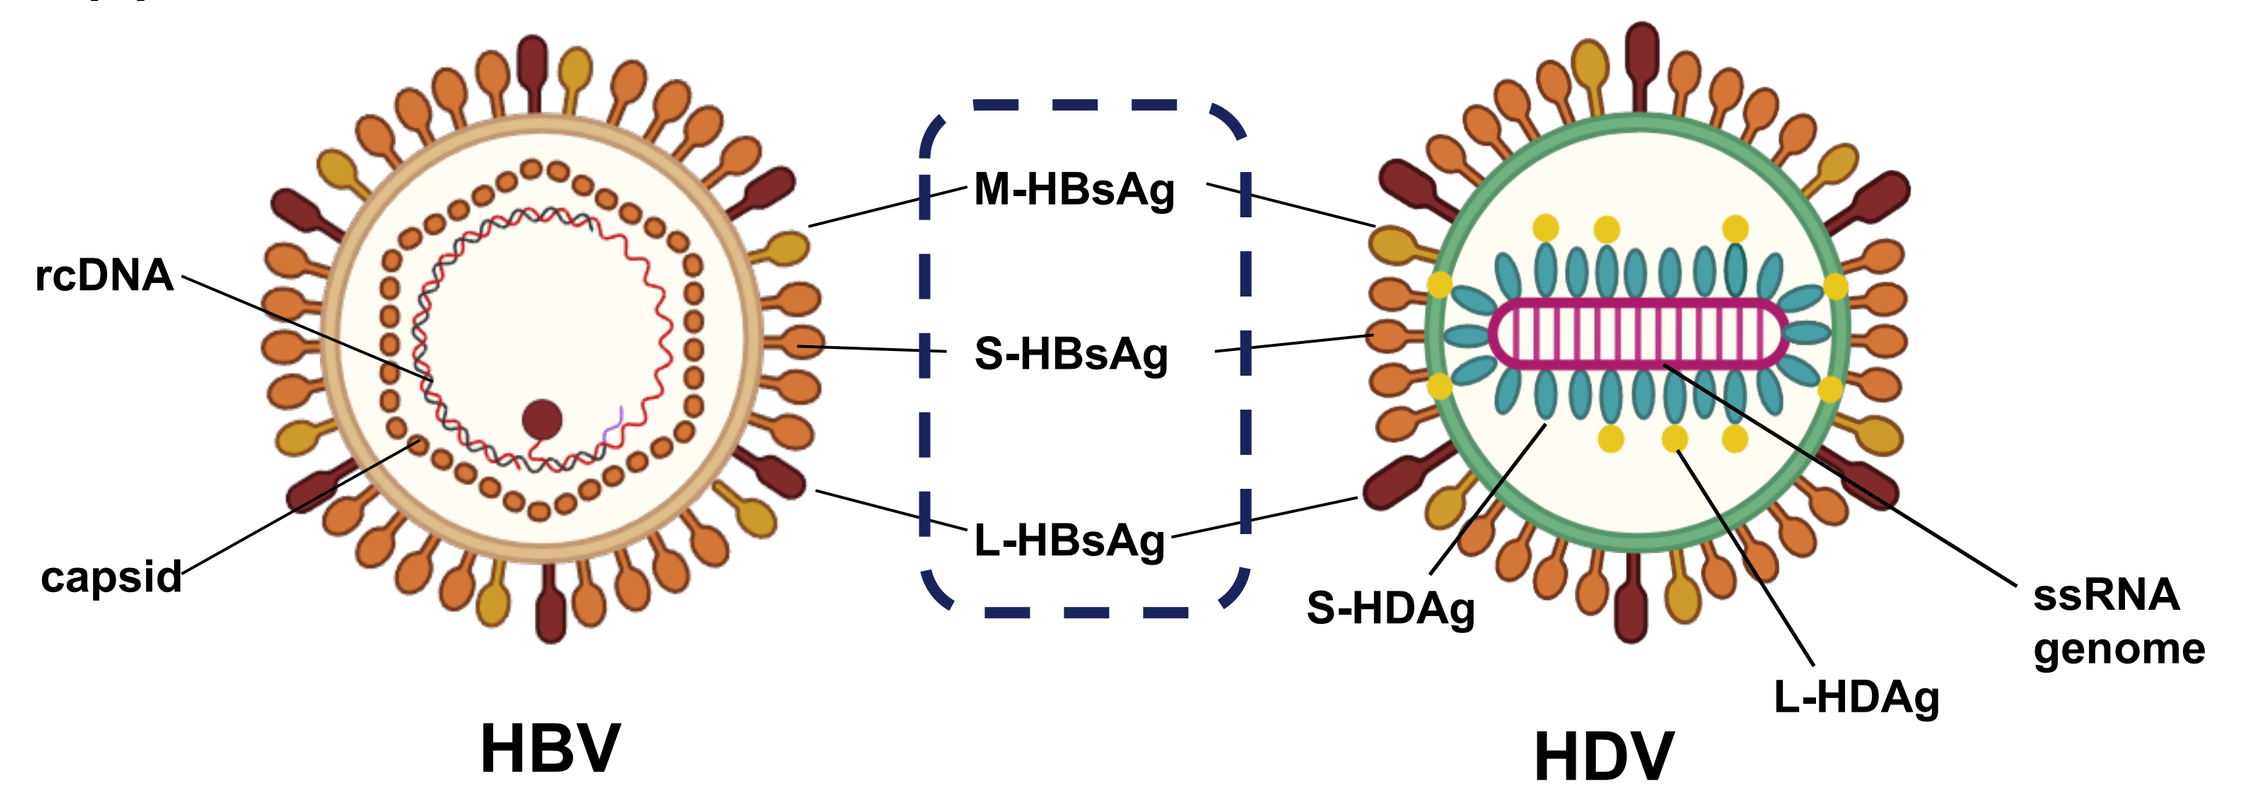
rcDNA
capsid
M-HBsAg
S-HBsAg
L-HBsAg
S-HDAg
L-HDAg
ssRNA genome
HBV
HDV

Sausen et al, 2022

Base editing

- Non dsDNA cleavage based technology
- Base editing leads to precise and permanent mutations in DNA

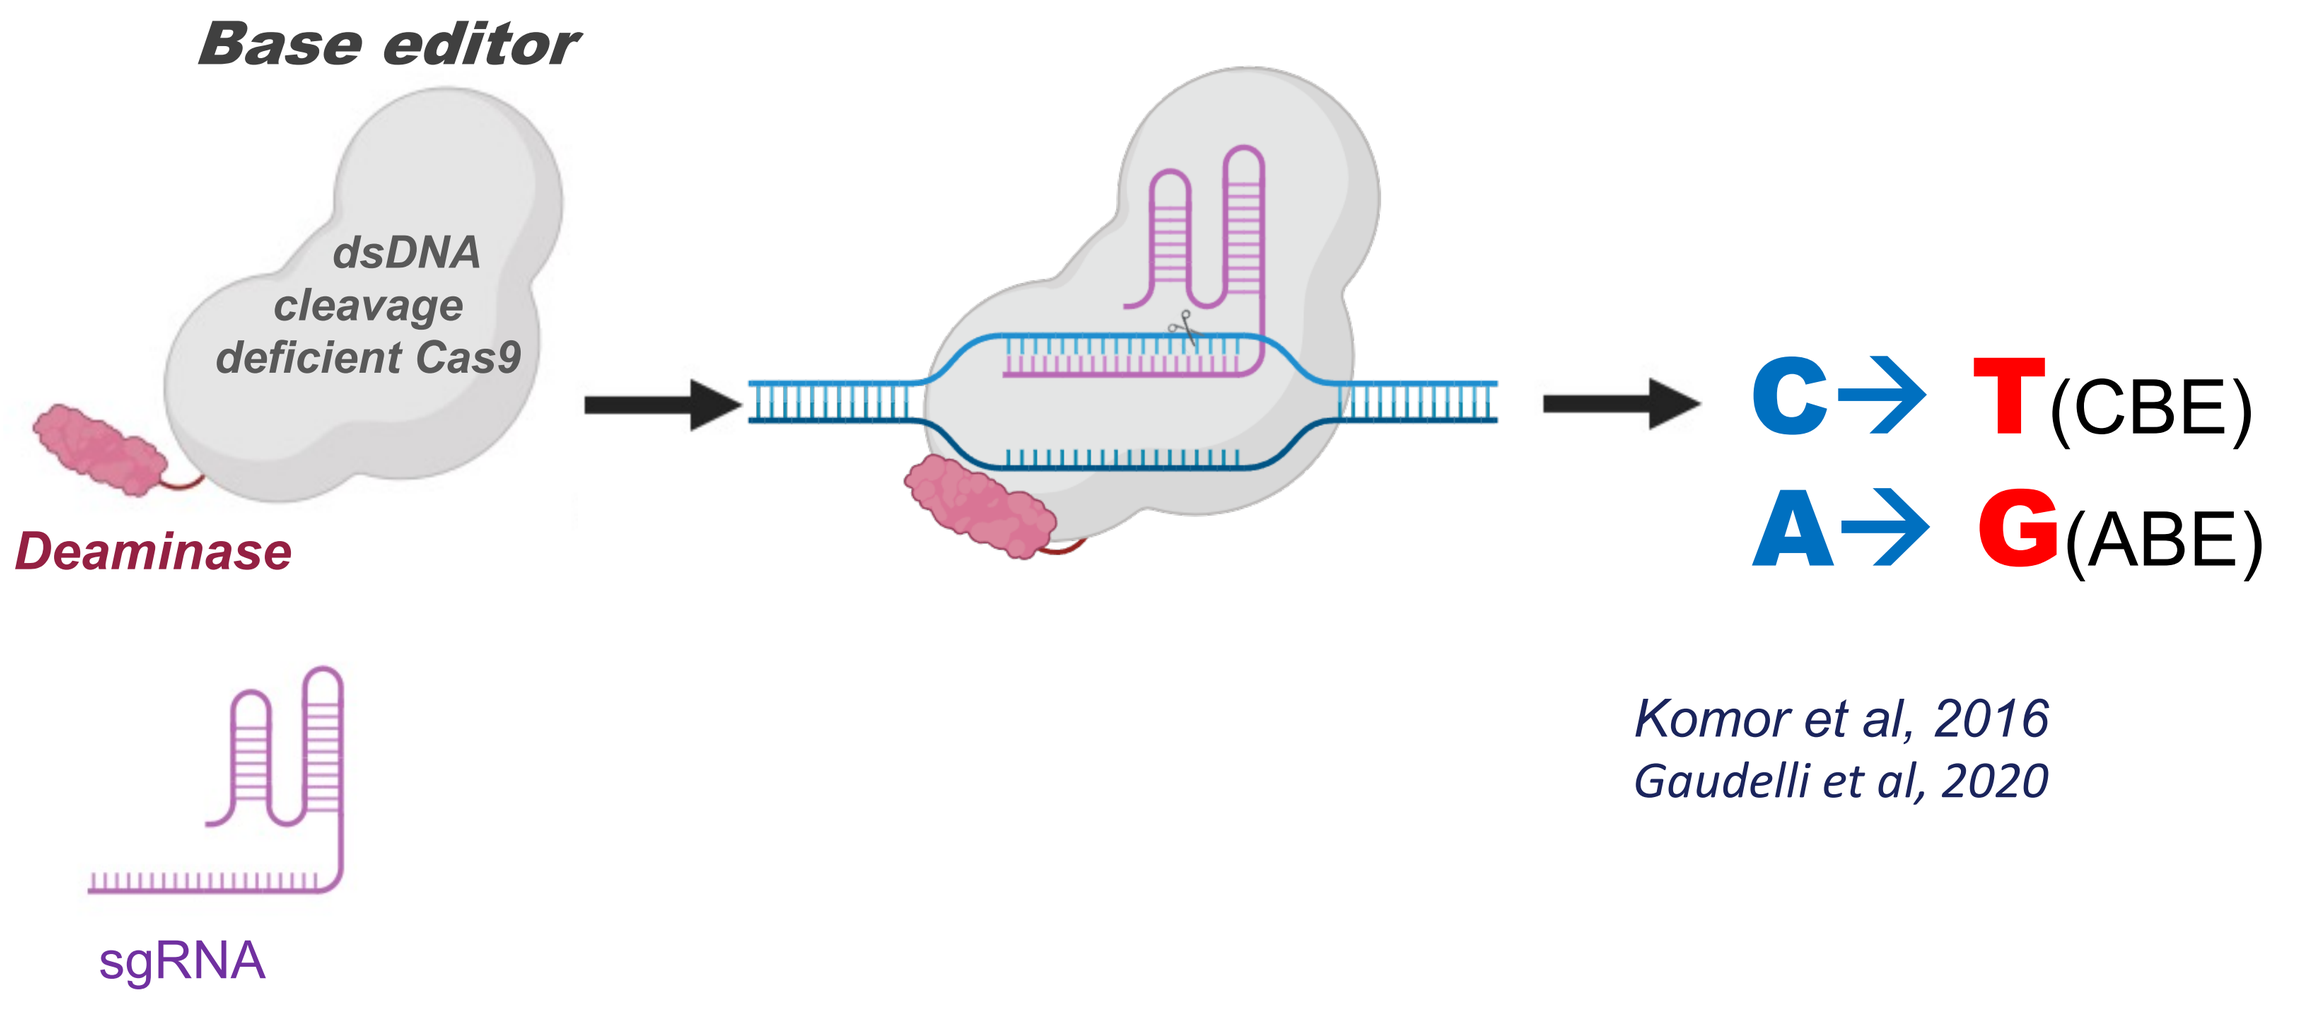
Base editor
dsDNA cleavage deficient Cas9
Deaminase
C→ T(CBE)
A→ G(ABE)
sgRNA

Komor et al, 2016
Gaudelli et al, 2020

Base editing & HBV

- Cytosine Base Editing (CBE) to functionally inactivate HBV genomes

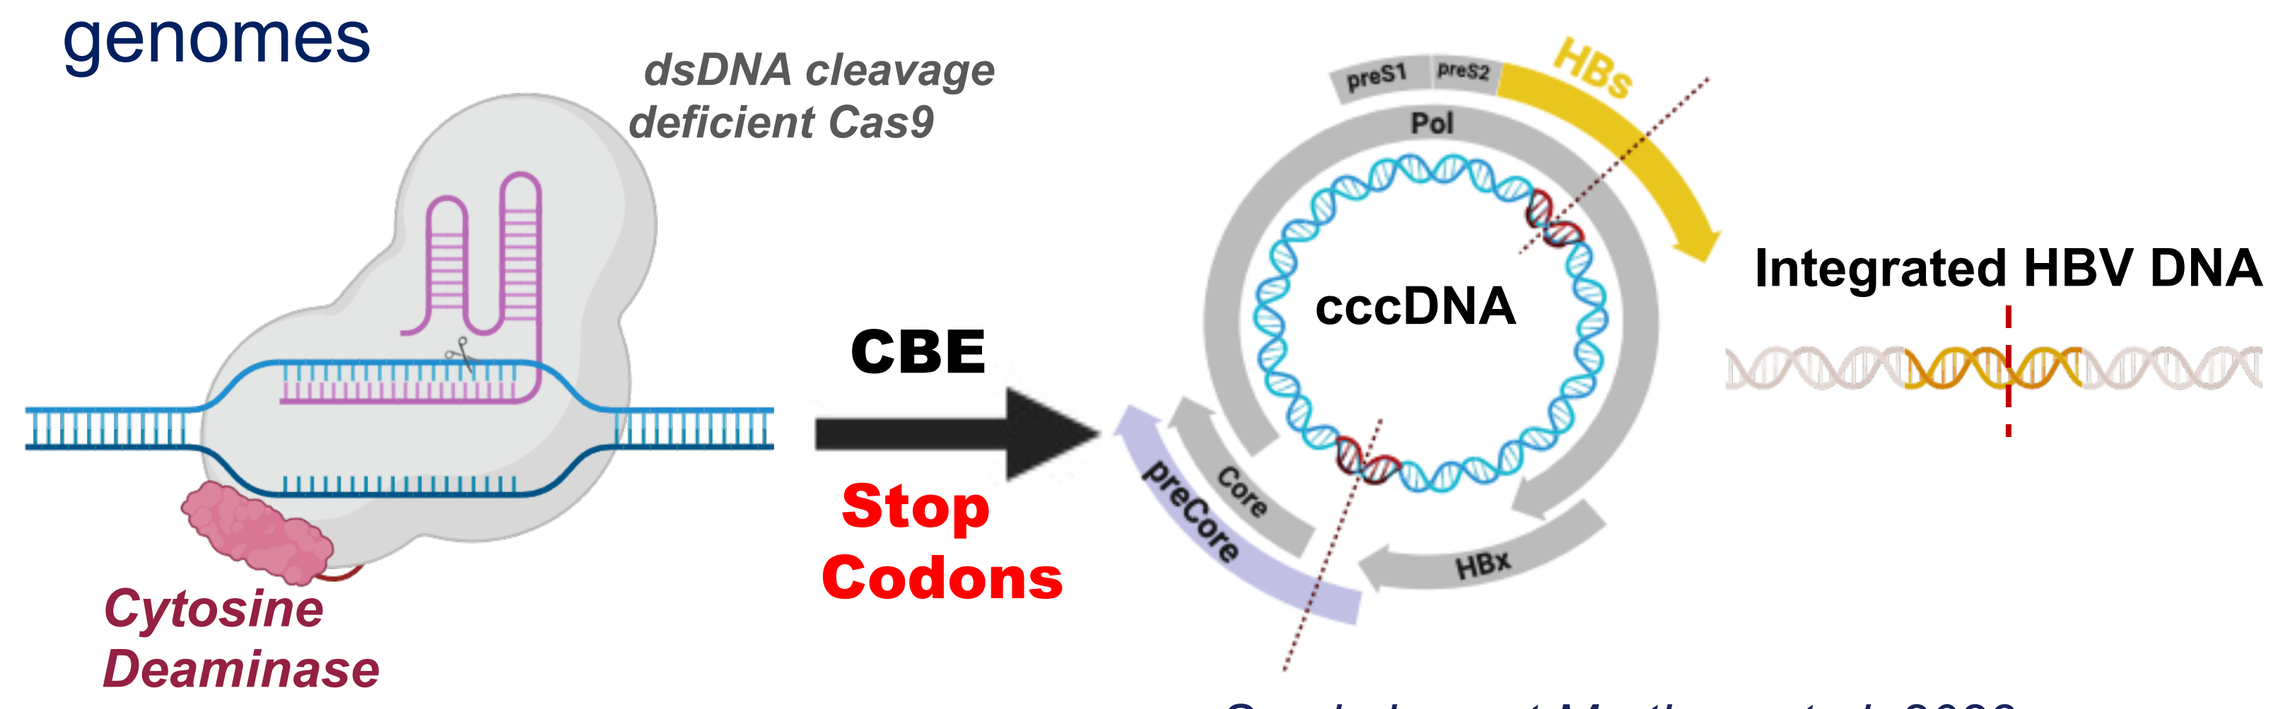
dsDNA cleavage deficient Cas9
Cytosine Deaminase
CBE
Stop Codons
cccDNA
Integrated HBV DNA

Smekalova et Martinez et al, 2023

- Adenine Base Editing (ABE) for sustained HBsAg reduction

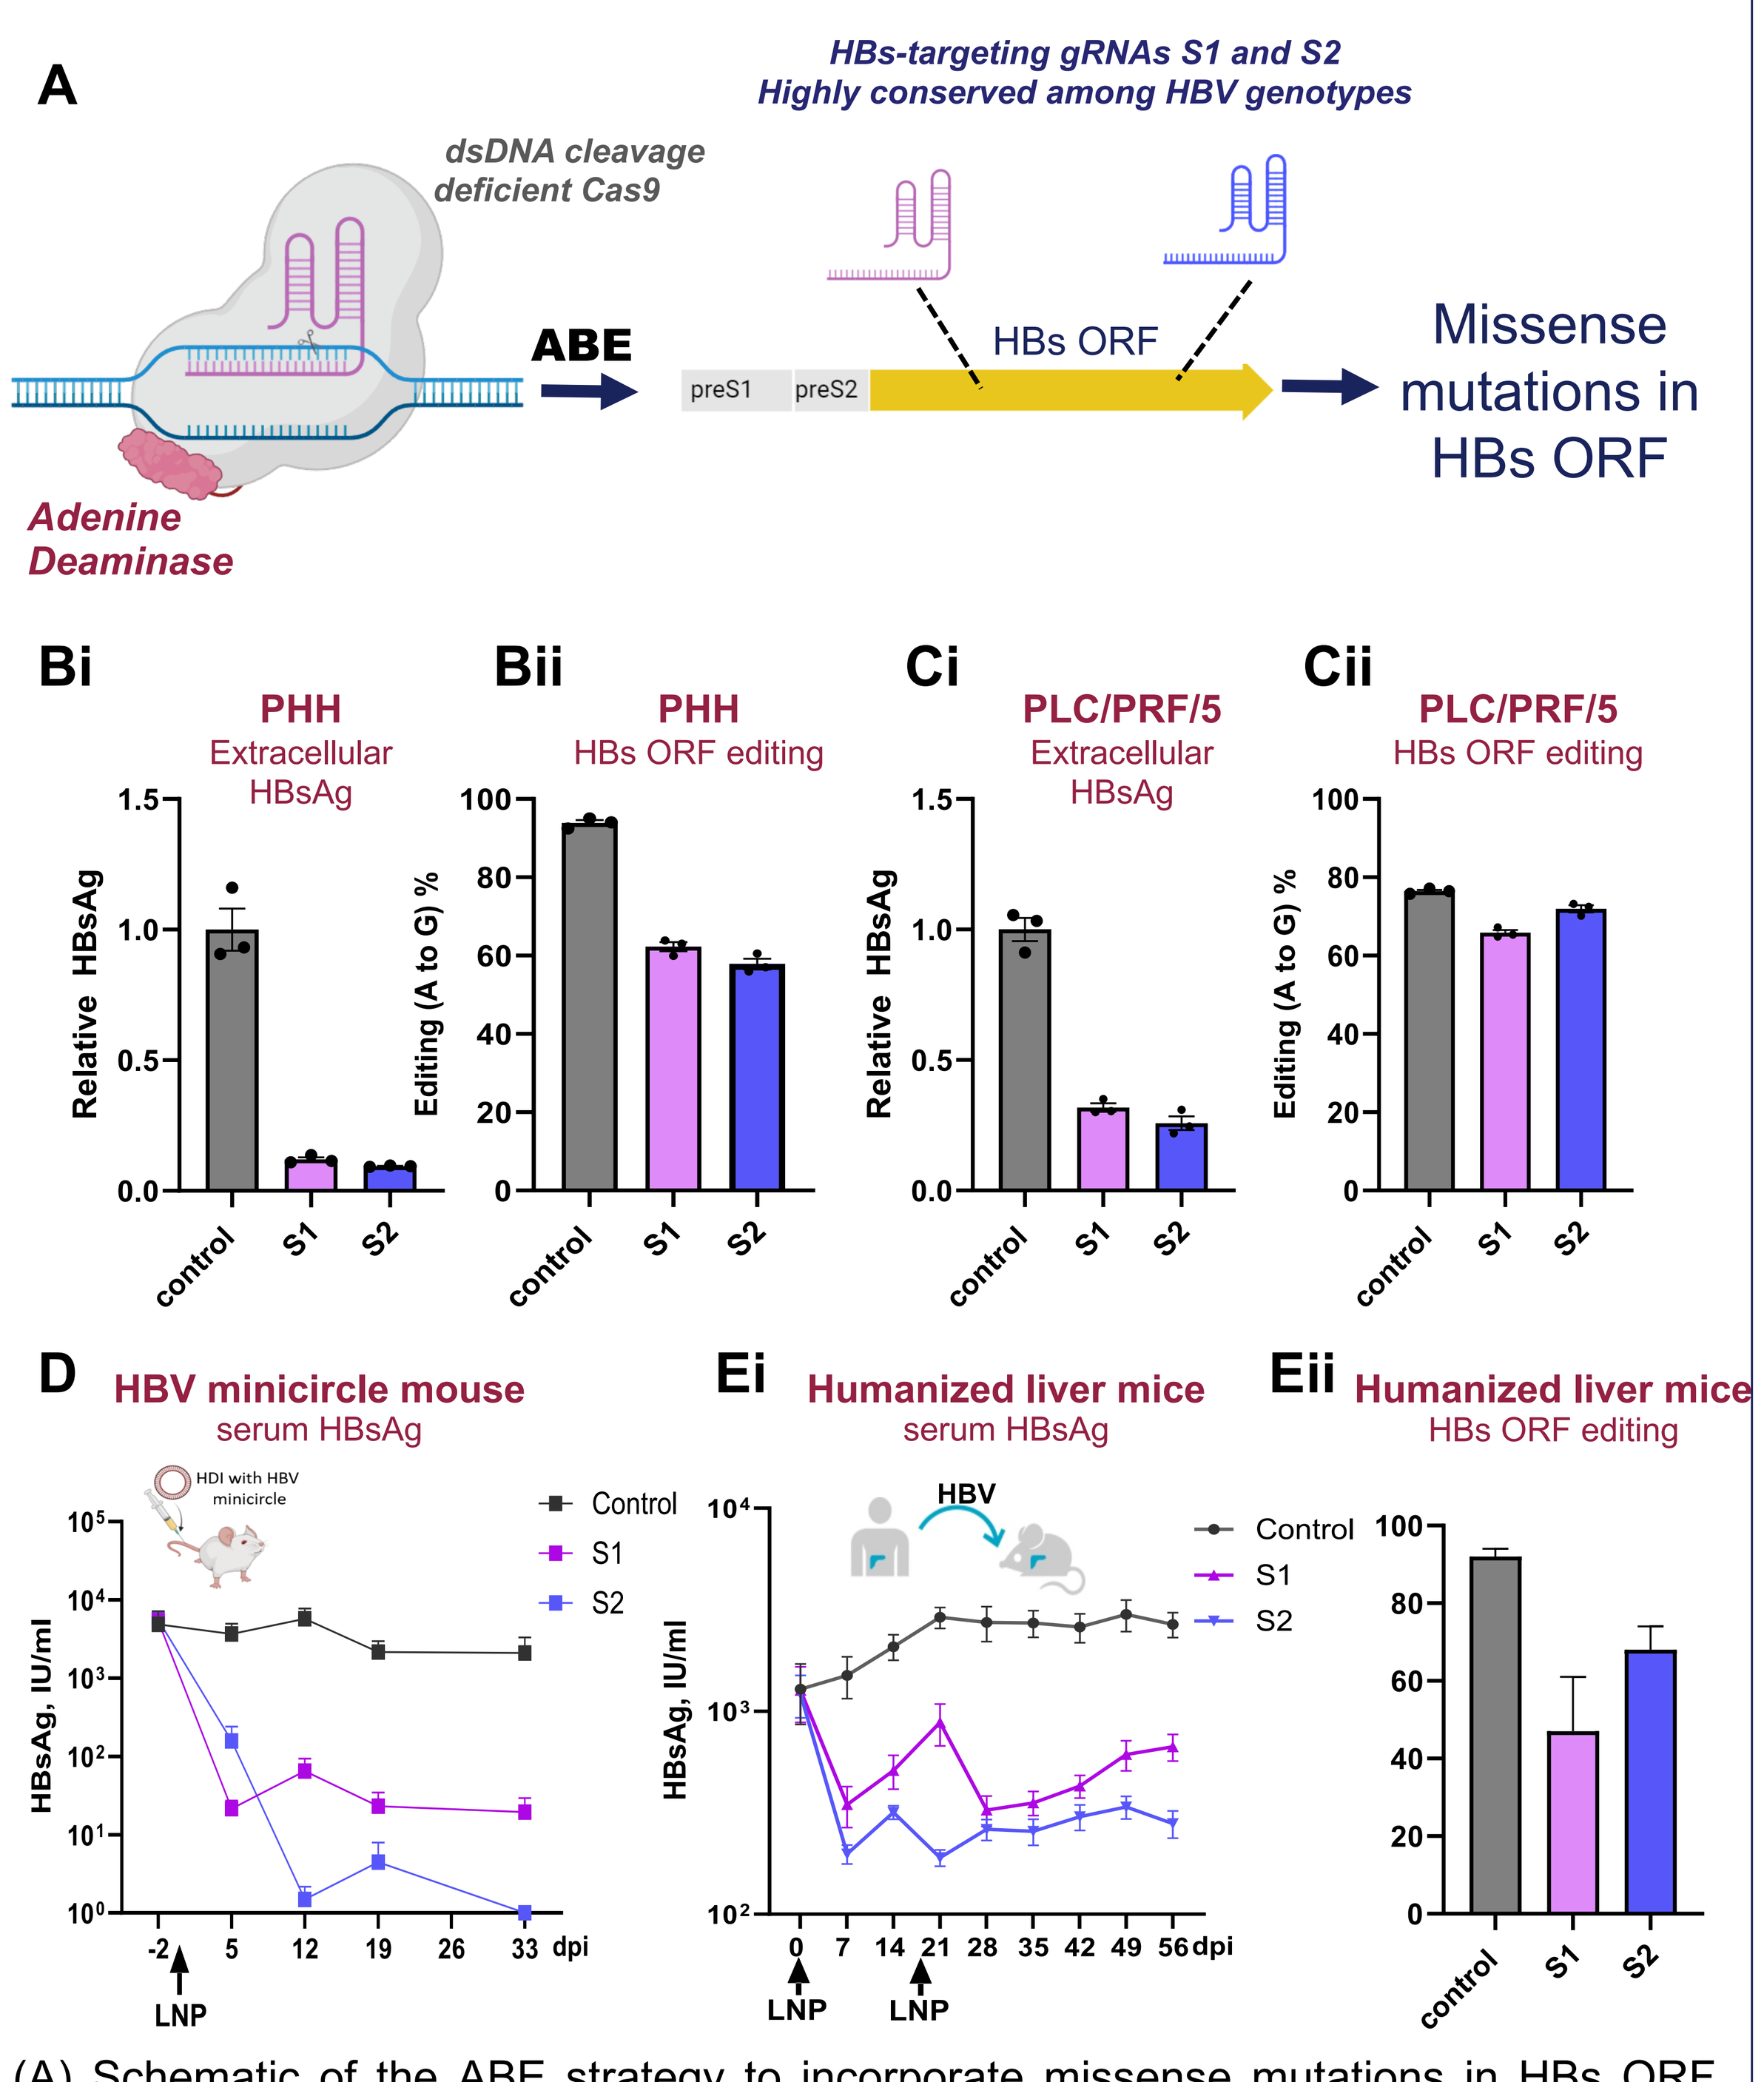
A
HBs-targeting gRNAs S1 and S2
Highly conserved among HBV genotypes
dsDNA cleavage deficient Cas9
Adenine Deaminase
ABE
preS1
preS2
HBs ORF
Missense mutations in HBs ORF
Bi PHH Extracellular HBsAg
Bii PHH HBs ORF editing
Ci PLC/PRF/5 Extracellular HBsAg
Cii PLC/PRF/5 HBs ORF editing
D HBV minicircle mouse serum HBsAg
Ei Humanized liver mice serum HBsAg
Eii Humanized liver mice HBs ORF editing

(A) Schematic of the ABE strategy to incorporate missense mutations in HBs ORF utilizing two gRNAs S1 and S2. (B-E) Effect of gRNA S1 and S2 on HBsAg levels in different *in vitro* and *in vivo* models. The editing efficiency on HBs ORF was determined by amplicon sequencing/NGS.

Aim of the study

To explore the potential of adenine base editing of HBs ORF in inhibiting HDV

Methods

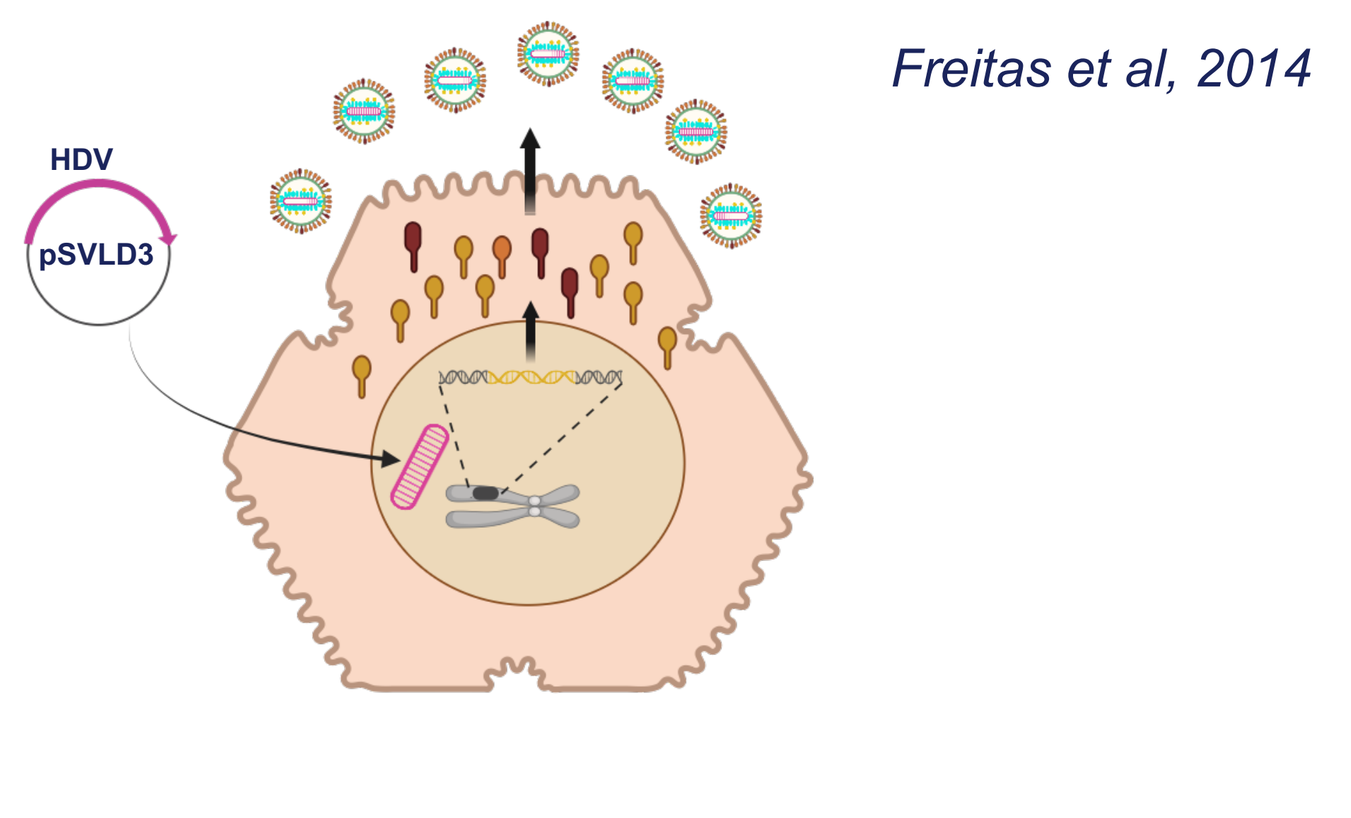
HDV
pSVLD3

Freitas et al, 2014

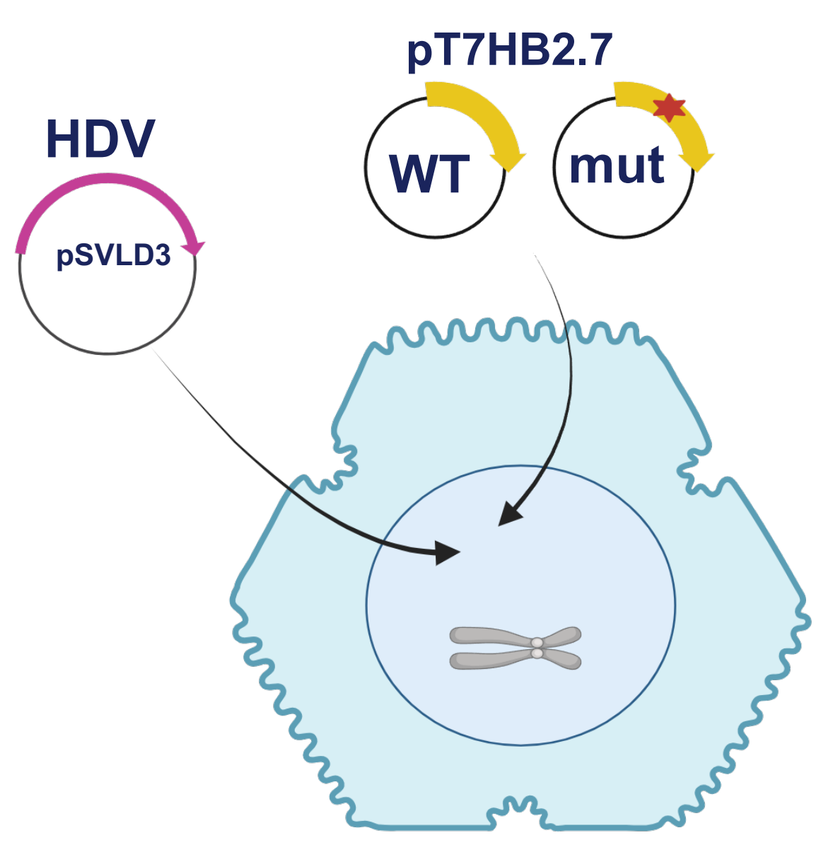
HDV
pSVLD3
pT7HB2.7
WT
mut

Base editing

- PLC/PRF/5 cells harboring replication incompetent integrated HBV DNA sequences capable of producing HBsAg. These cells support assembly and release of infectious HDV particles.
- HDV replication initiation was performed by transfecting PLC/PRF/5 cells with an HDV-encoding plasmid, pSVLD3.
- ABE was performed by transfecting PLC/PRF/5 cells with an ABE encoding mRNA and gRNA S1 or S2.
- **Base Editing followed by HDV replication (Results Panel I)**
- **HDV replication followed by Base Editing (Results Panel II)**

Transfection

- Huh7 cells were used to perform plasmid transfection.
- gRNA S2 corresponding mutations were incorporated in HBs-encoding plasmid, pT7HB2.7.
- Huh7 cells were co-transfected with pSVLD3 and either WT or mutant pT7HB2.7.
- **Cotransfection: pSVLD3 and HBs-encoding WT/mutant (Results Panel III)**

Readout : **Assessement of HBsAg and extracellular HDV RNA**

Results

I - Base editing of HBs ORF inhibits HDV release

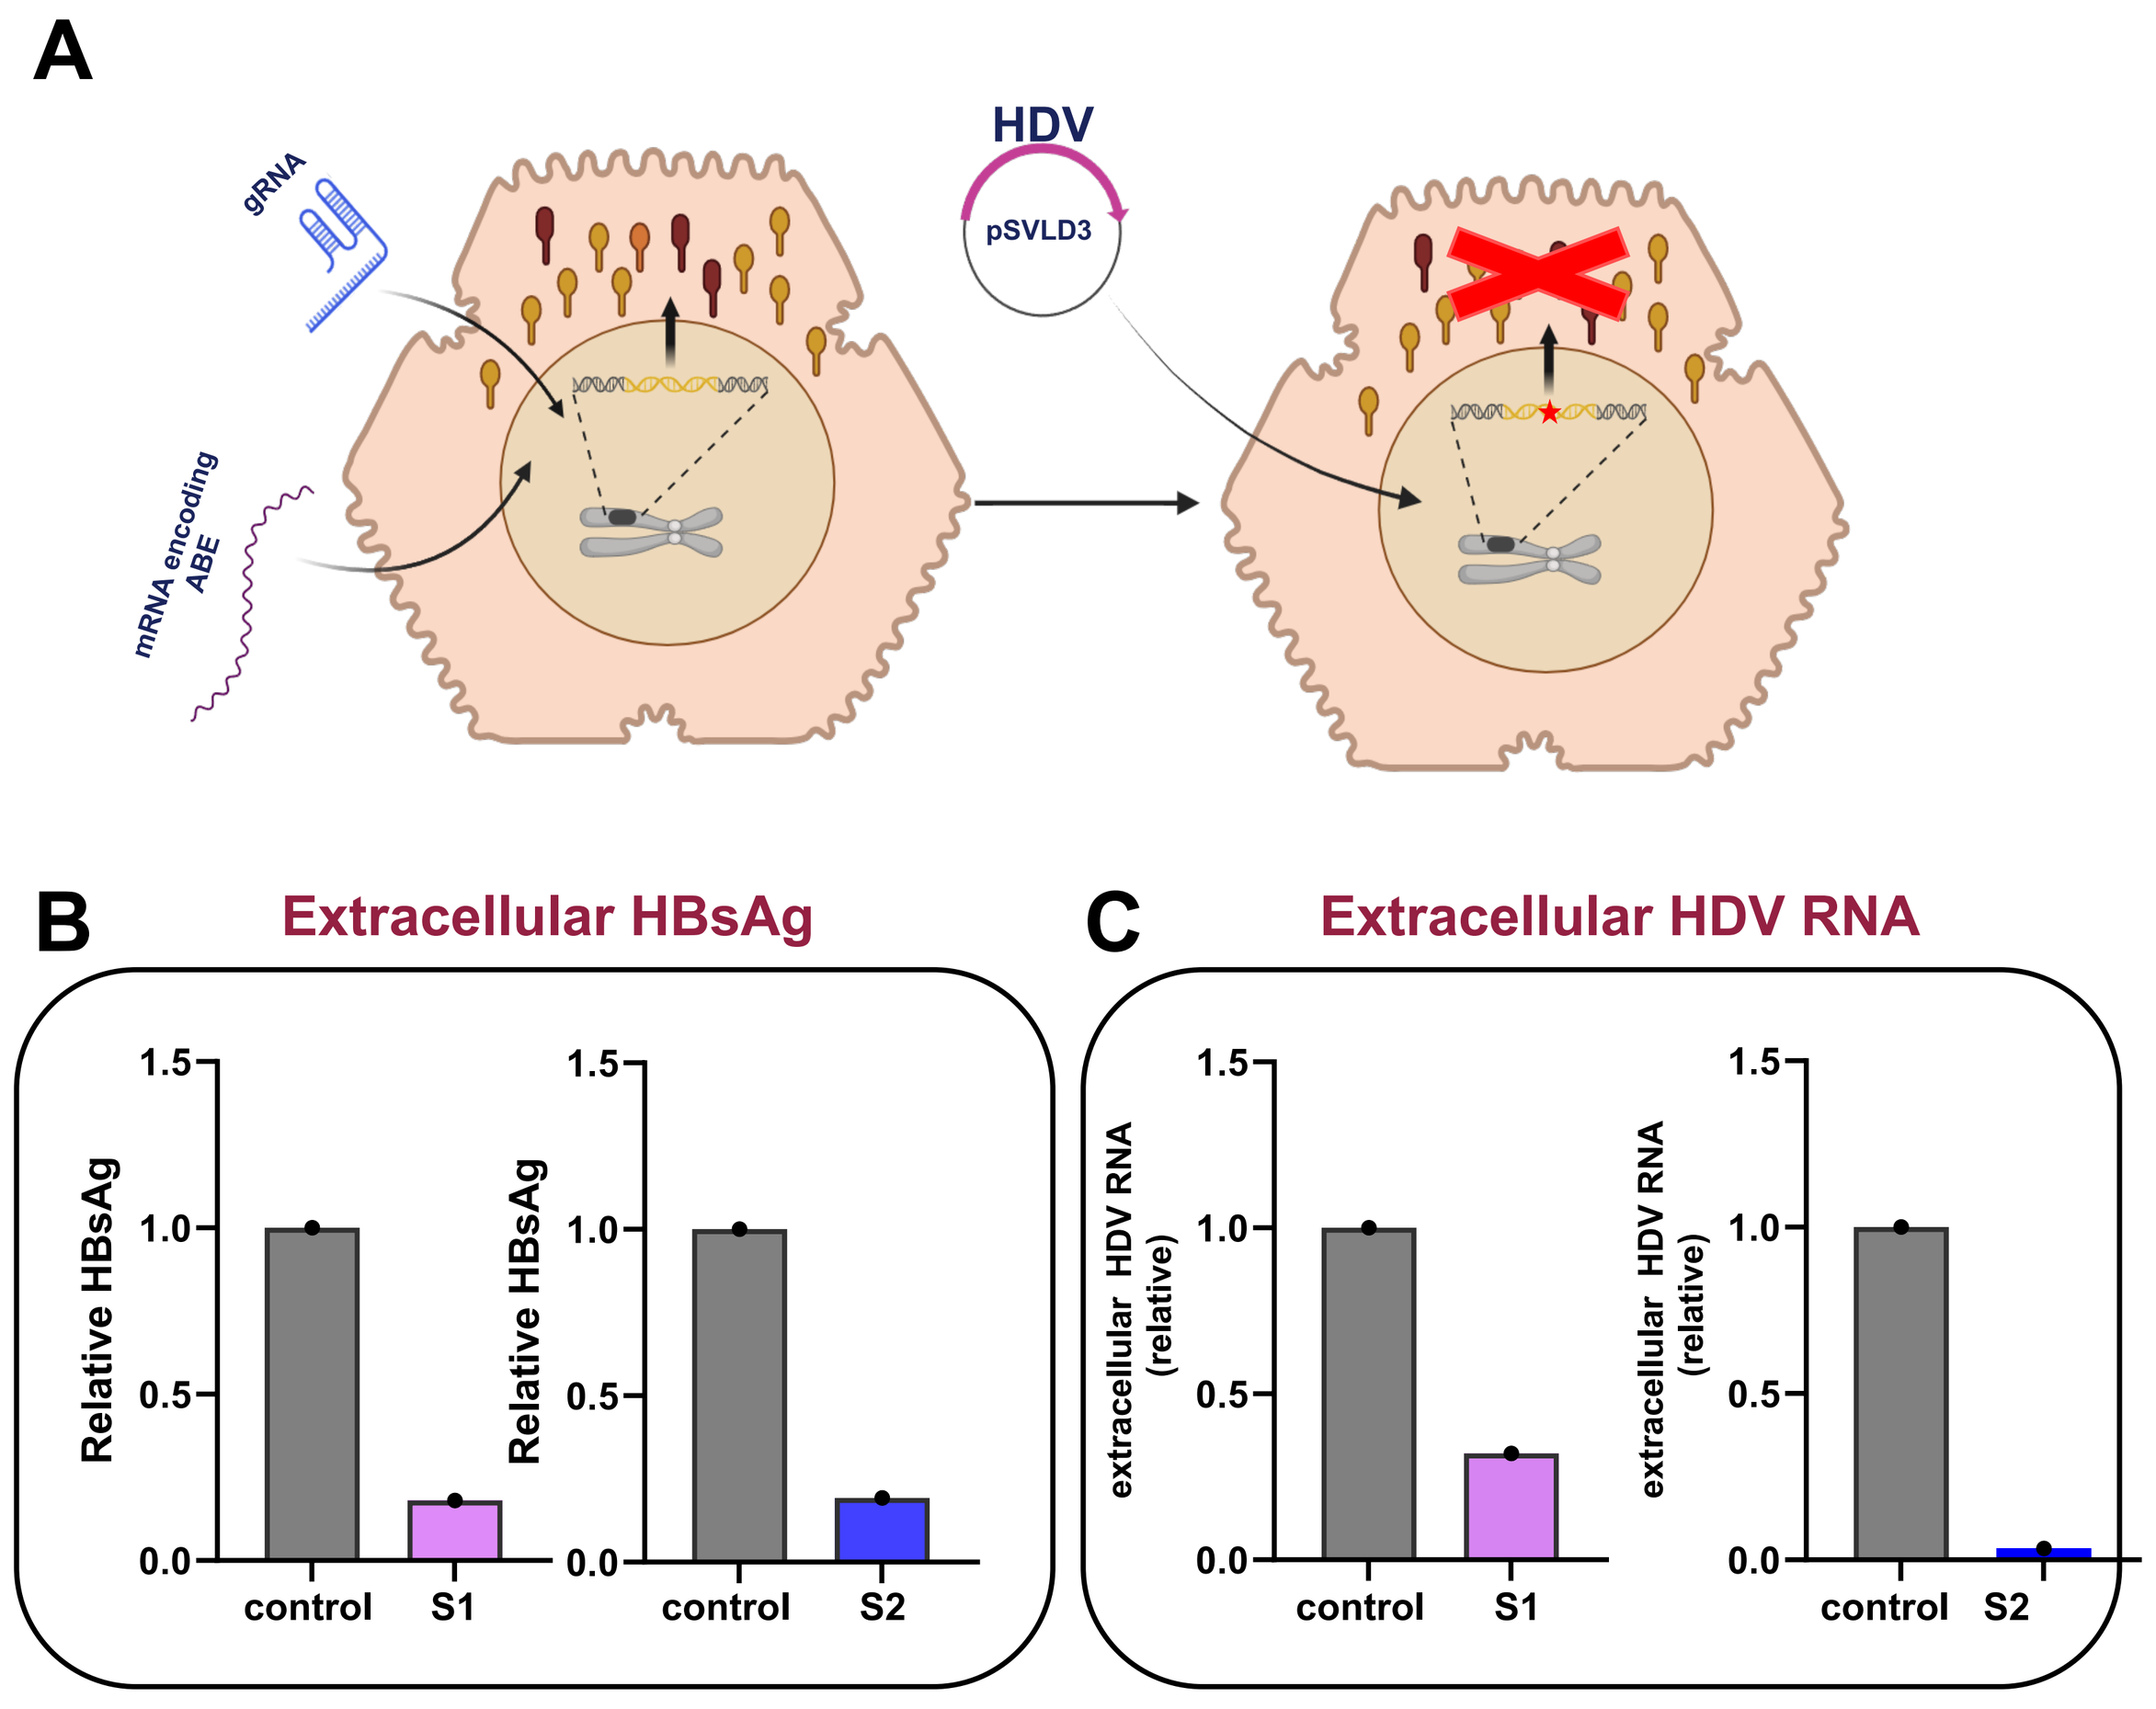
A
gRNA
mRNA encoding ABE
HDV
pSVLD3
B Extracellular HBsAg
C Extracellular HDV RNA
control
S1
S2

(A) Schematic of the experimental plan. PLC/PRF/5 cells were first transfected with ABE-encoding mRNA and gRNA (S1/S2) to perform base editing, followed by transfection with pSVLD3 5 days later. (B) At 15th day post-pSVLD3 transfection, levels of extracellular HBsAg were determined by ELISA and (C) extracellular HDV RNA was quantified by one-step qRT-PCR. Control represents non HBs-targeting gRNA.

II - Base editing of HBs ORF inhibits HDV release & spread

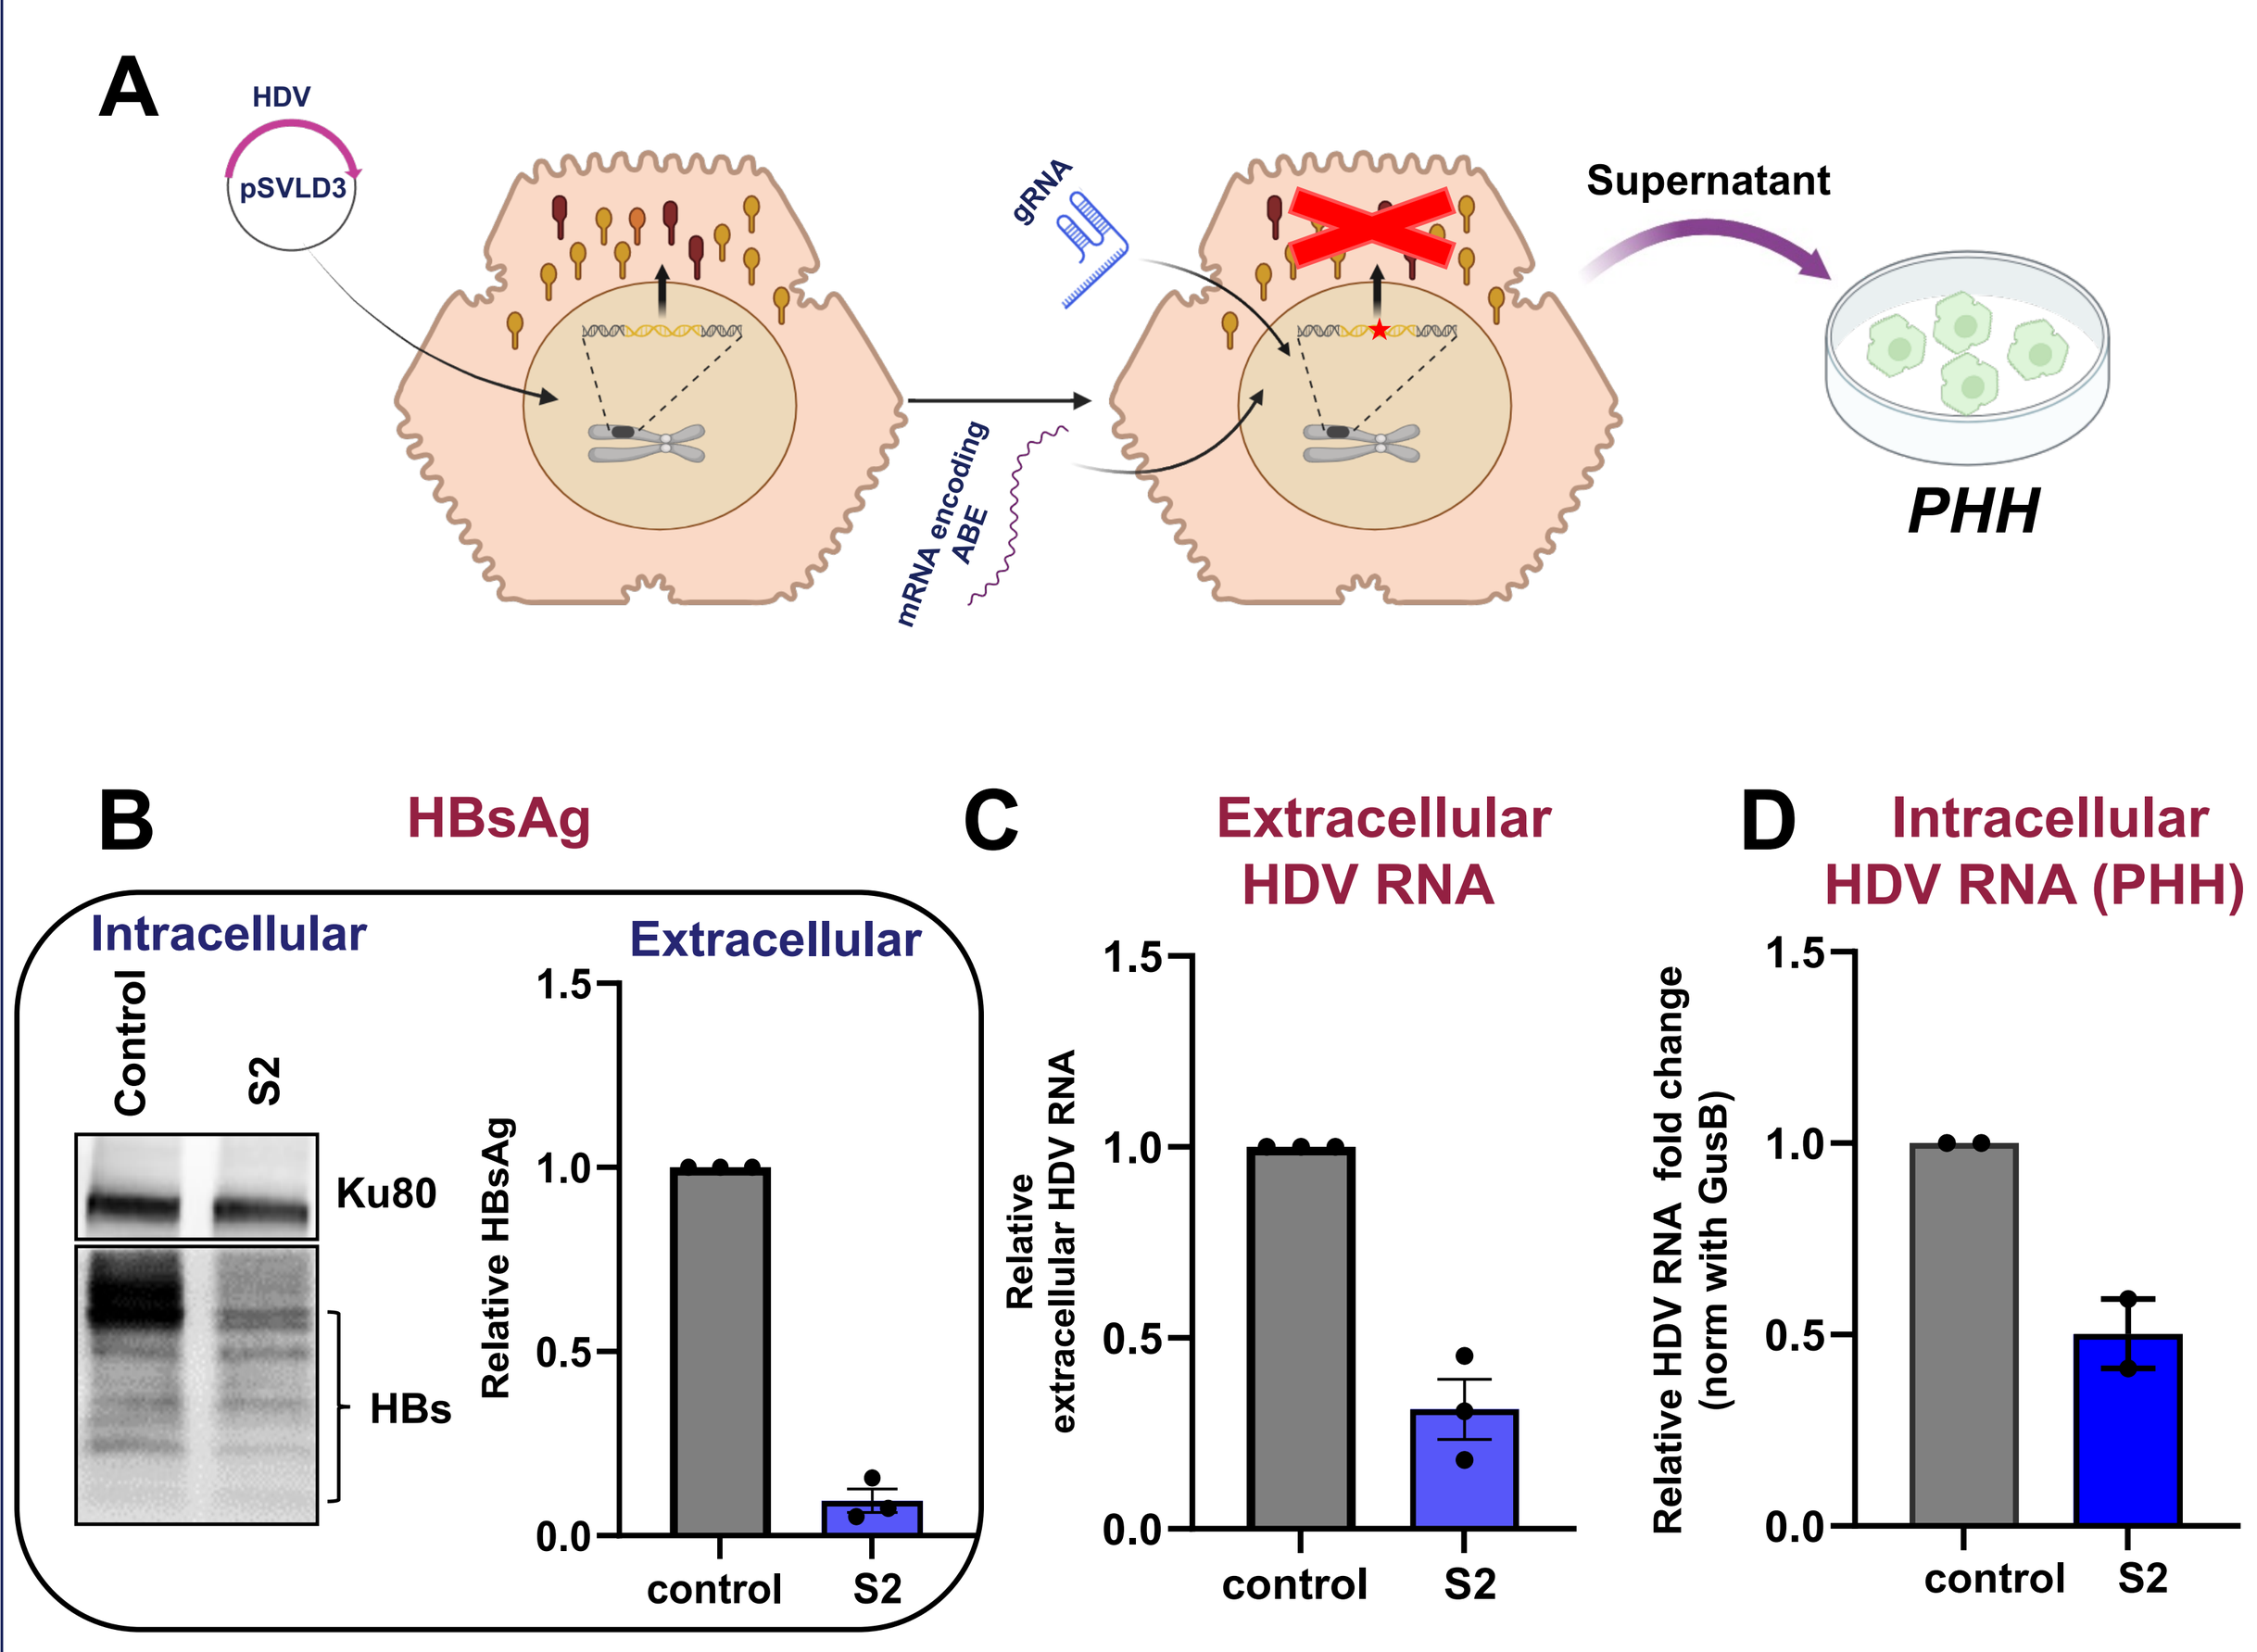
A
HDV
pSVLD3
gRNA
mRNA encoding ABE
Supernatant
PHH
B HBsAg
Intracellular
Extracellular
Ku80
HBs
C Extracellular HDV RNA
D Intracellular HDV RNA (PHH)
control
S2

(A) Schematic of the experimental plan. PLC/PRF/5 cells were first transfected with pSVLD3 followed by ABE 5 days later (B) At 11th day post pSVLD3 transfection, levels of intracellular and extracellular HBsAg were determined by Western blot and ELISA, respectively. Ku80 served as internal control of loading. (C) Extracellular HDV RNA was quantified by one-step qRT-PCR. (D) PHH were infected with equal volume of HDV released from control gRNA or gRNA S2-edited PLC/PRF/5 cells. At 6th day post infection, levels of intracellular HDV RNA were quantified by one-step qRT-PCR.

III - HBs ORF mutation abrogates HDV release and PHH infectivity

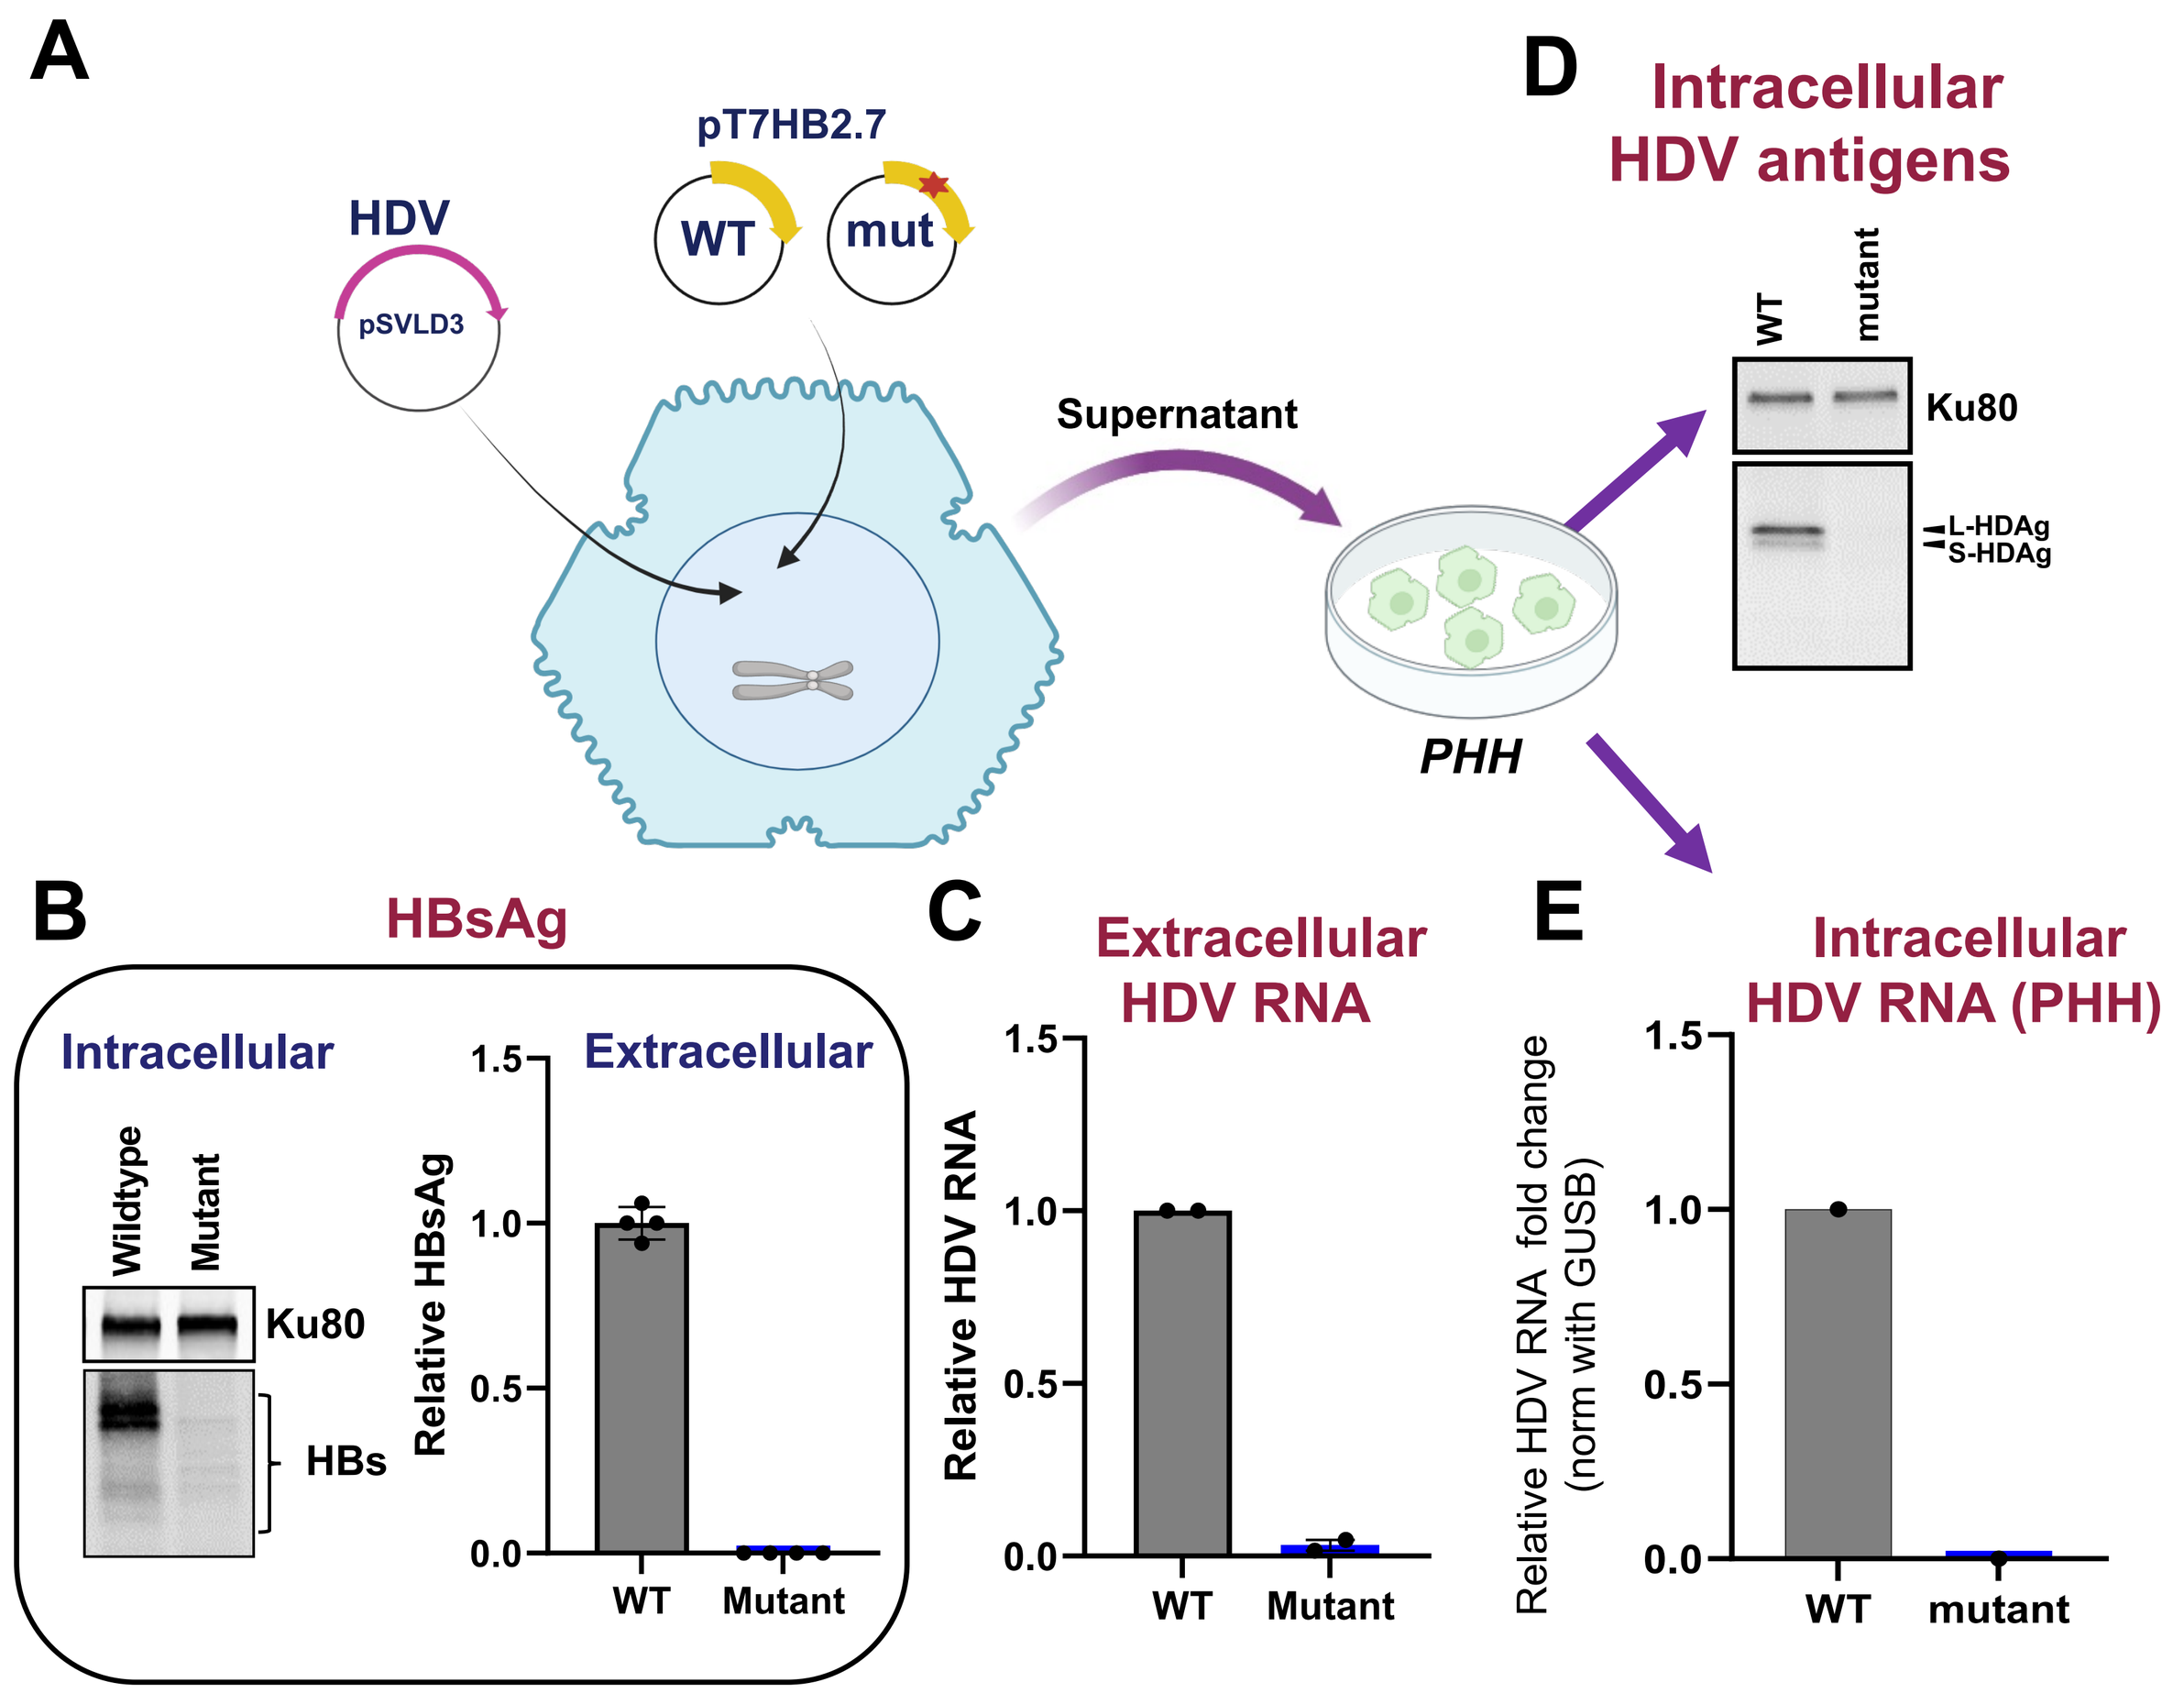
A
HDV
pSVLD3
pT7HB2.7
WT
mut
Supernatant
PHH
D Intracellular HDV antigens
Ku80
L-HDAg
S-HDAg
B HBsAg
Intracellular
Extracellular
Wildtype
Mutant
HBs
C Extracellular HDV RNA
E Intracellular HDV RNA (PHH)

(A) Schematic of the experimental plan. Huh7 cells were co-transfected with pSVLD3 and wildtype (WT) or gRNA S2 corresponding mutant. (B) At 11th day post transfection, intracellular and extracellular HBsAg levels were determined by Western blot and ELISA, respectively. (C) Extracellular HDV RNA was quantified by one-step qRT-PCR. (D-E) PHH were infected with equal volume of HDV released from Wildtype (WT) or mutant conditions. At 6th day post infection, intracellular HDAg levels and intracellular HDV RNA levels were determined by Western blot and one-step qRT-PCR, respectively.

Conclusions

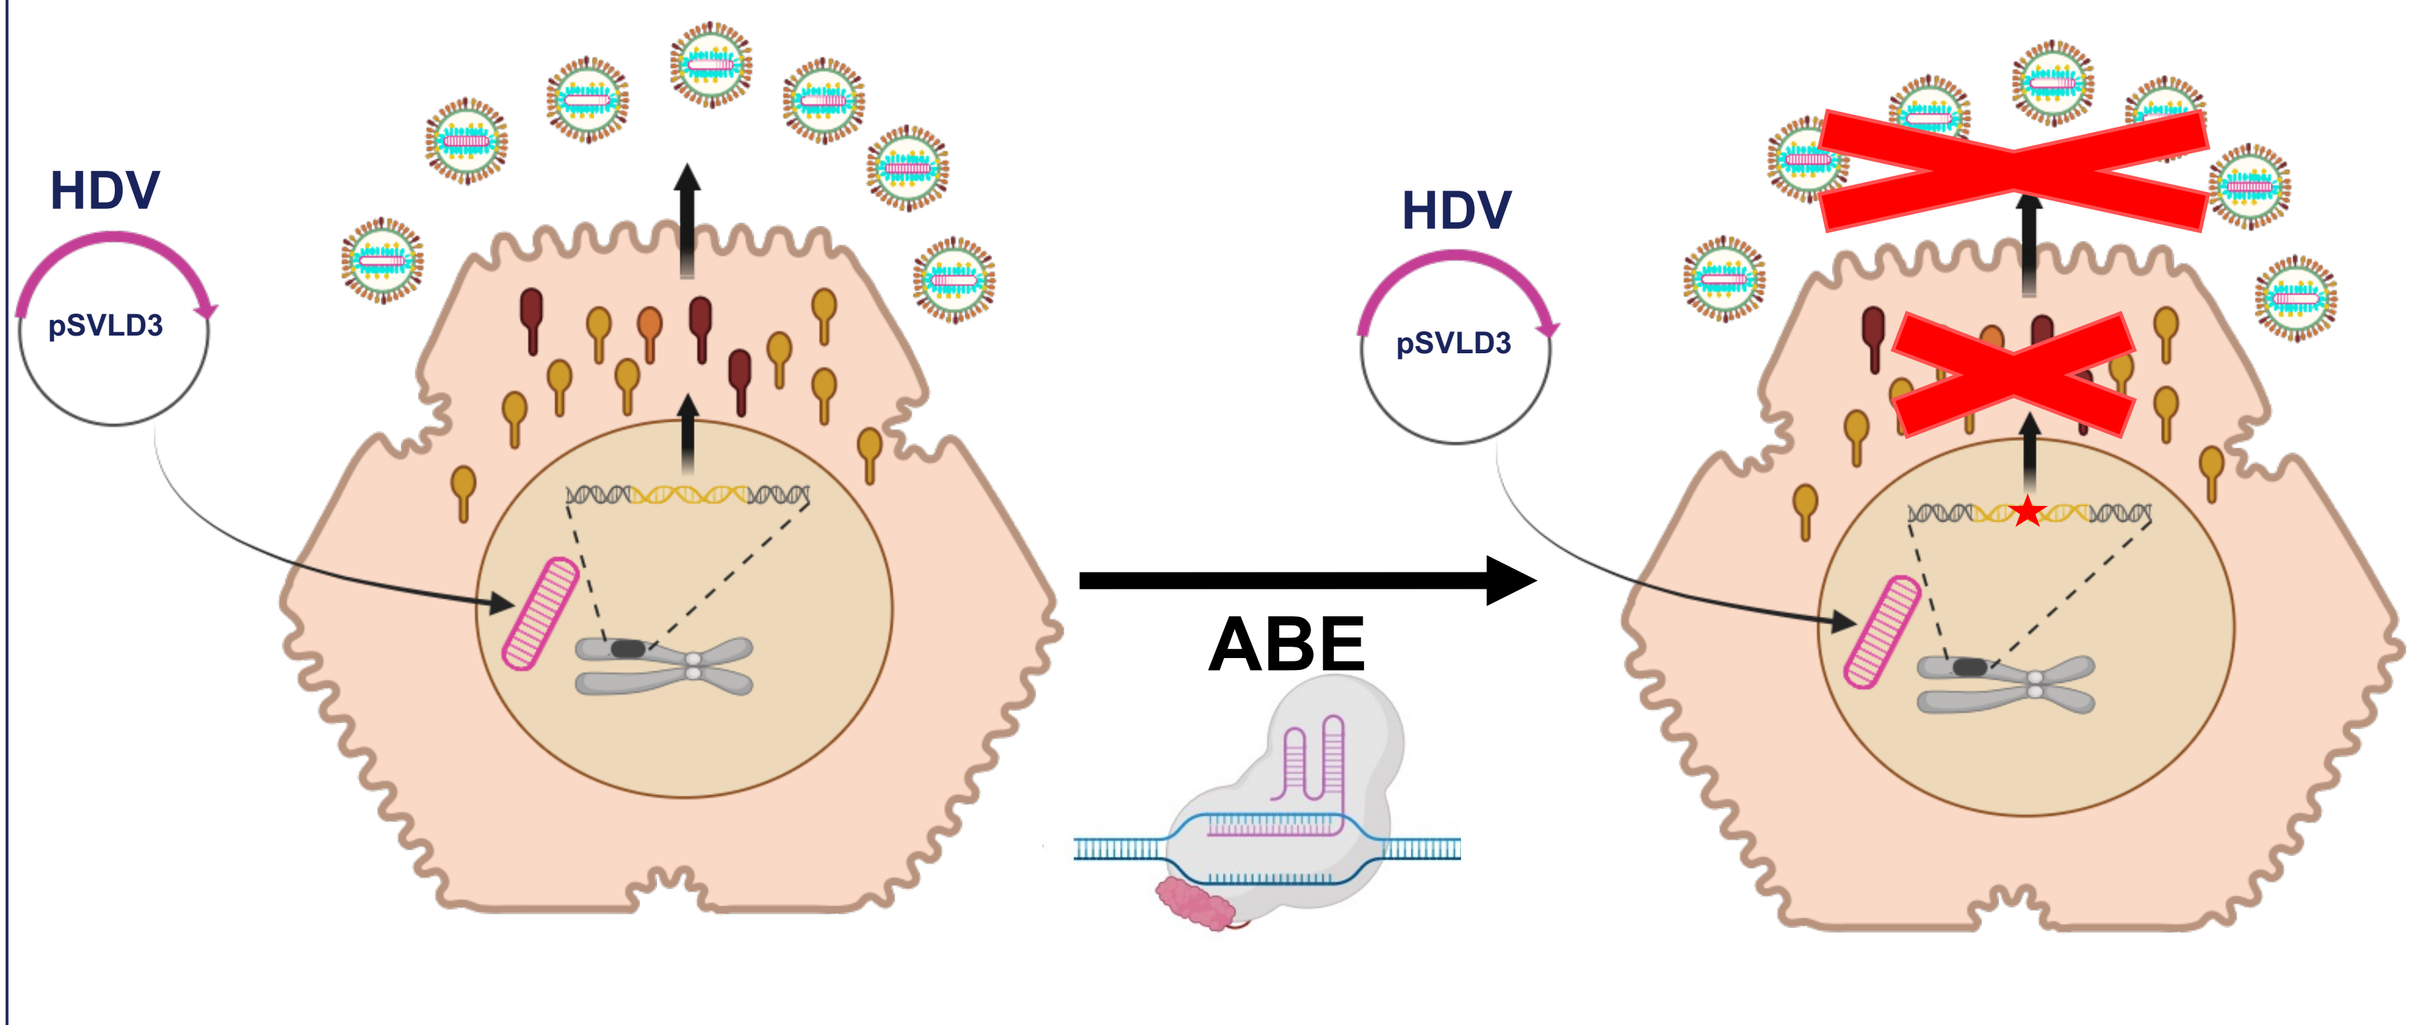
HDV
pSVLD3
ABE

Adenine base editing is a promising approach to mutate HBs ORF to :

- **inhibit HBsAg expression from cccDNA and integrated HBV DNA**
- **potently reduce HDV release and spread**

Acknowledgements

Inserm · Beam Therapeutics · Lyon Hepatology Institute · cnrs · CRCL · HCL · Centre Léon Bérard · Lyon 1

References

1. Sausen DG, et al. Hepatitis B and Hepatitis D Viruses: A Comprehensive Update with an Immunological Focus. Int J Mol Sci. 2022 Dec 15;23(24):15973.
2. Komor AC,et al. Programmable editing of a target base in genomic DNA without double-stranded DNA cleavage. Nature. 2016 May 19;533(7603):420-4.
3. Gaudelli NM, et al. Directed evolution of adenine base editors with increased activity and therapeutic application. Nat Biotechnol. 2020 Jul;38(7):892-900.
4. Smekalova EM, Martinez MG, et al. Cytosine base editing inhibits hepatitis B virus replication and reduces HBsAg expression *in vitro* and *in vivo*. Mol Ther Nucleic Acids. 2023 Dec 27;35(1):102112.
5. Freitas N, et al. Envelope proteins derived from naturally integrated hepatitis B virus DNA support assembly and release of infectious hepatitis delta virus particles. J Virol. 2014 May;88(10):5742-54.

Disclosure

This work was funded by Beam Therapeutics, a public company developing base editing technology for human therapeutics. E. Smekalova, S. Dejene, M. Packer, C-Y Chen, G. Ciaramella, are/were employees/shareholders of Beam Therapeutics.